C3d-Directed Factor H Targeting Delivers Potent and Durable Complement Inhibition and Disease-Modifying Efficacy In Kidney and Skin Without Inhibiting Systemic Complement

Fei Liu[1], Sarah Ryan[1], Kelly Fahnoe[1], Jennifer Morgan[1], Katja Bieber[2], Enno Schmidt[2], Admar Verschoor[2], Ralf J Ludwig[2], David J Salant[3], Joshua M Thurman[4], V Michael Holers[4], Shelia Violette[1], Stefan Wawersik[1]

[1] Q32 Bio; [2] University of Lübeck; [3] Boston University School of Medicine; [4] University of Colorado Denver School of Medicine

Background

While systemic complement inhibition is a therapeutic strategy for complement-driven diseases, potency and durability of inhibition are limited by high circulating concentrations and rapid turnover of complement proteins. Additionally, because of complement's essential role in innate and adaptive immunity, systemic blockade raises infection risk in patients. As a consequence, substantial unmet need remains for safer and more effective anti-complement therapies, particularly for chronic diseases. We hypothesized that C3d-mediated localization of a catalytic inhibitor of convertase function can potently inhibit local complement activation in diseased tissue while minimizing systemic blockade. To test this, we designed ADX-097, a bi-functional fusion protein comprising a humanized anti-C3d monoclonal antibody linked to two moieties of the first five consensus repeats of factor H (fH_{1-5})(**Figure 1**).

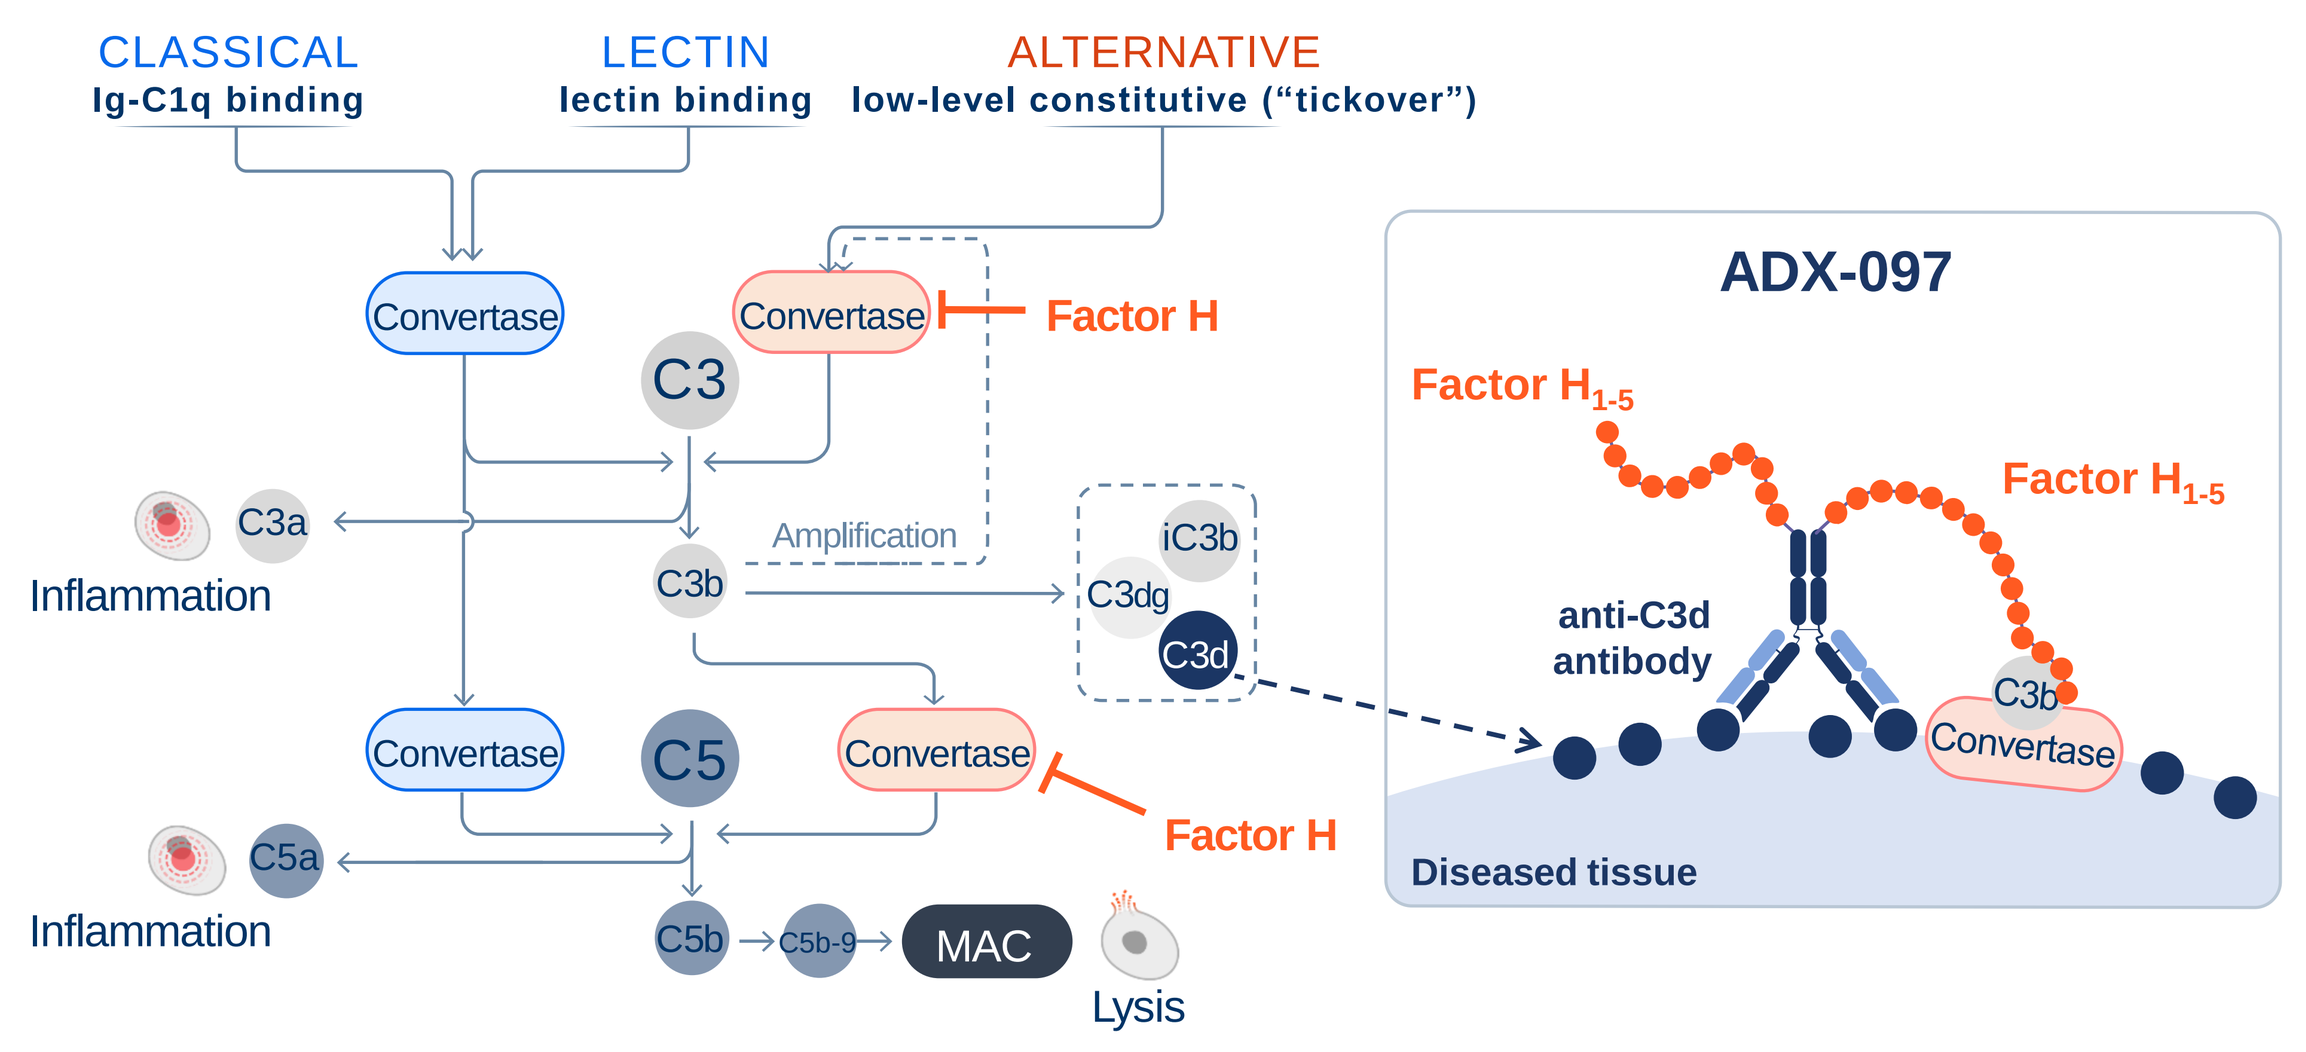
CLASSICAL
Ig-C1q binding
LECTIN
lectin binding
ALTERNATIVE
low-level constitutive ("tickover")
Convertase
Factor H
C3
C3a
Inflammation
C3b
Amplification
iC3b
C3dg
C3d
C5
C5a
C5b
C5b-9
MAC
Lysis
ADX-097
Factor H1-5
anti-C3d antibody
Diseased tissue

Figure 1. ADX-097 Targets AP Complement Inhibition to Tissues with High-Density C3d Deposition. ADX-097 is a bi-functional fusion protein that combines an anti-C3d monoclonal antibody with two moieties of the first five SCRs of factor H (fH_{1-5}). ADX-097 is designed to preferentially bind to tissues with active AP convertase activity, where C3d is locally deposited at high density. C3d binding brings the fH_{1-5} domains into proximity of the AP convertase, where fH_{1-5} catalyzes dissociation and degradation (decay acceleration) of the convertase complex.

ADX-097 Potently Ameliorates Proteinuria in Passive Heymann Nephritis

Study Design:
Disease induction on Day 0
ADX-097 treatment on Day 3
Tissue collection, Day 5

All Tested Doses of ADX-097 Inhibit Proteinuria
Urine protein:creatinine ratio

ADX-097 Inhibits Glomerular Complement Activity
Quantitation of anti-C3c IF in Day 5 kidneys

ADX-097 Doses ≤ 10 mg/kg Do Not Affect Systemic Complement
Measured as deposition of C3 on Zymosan beads

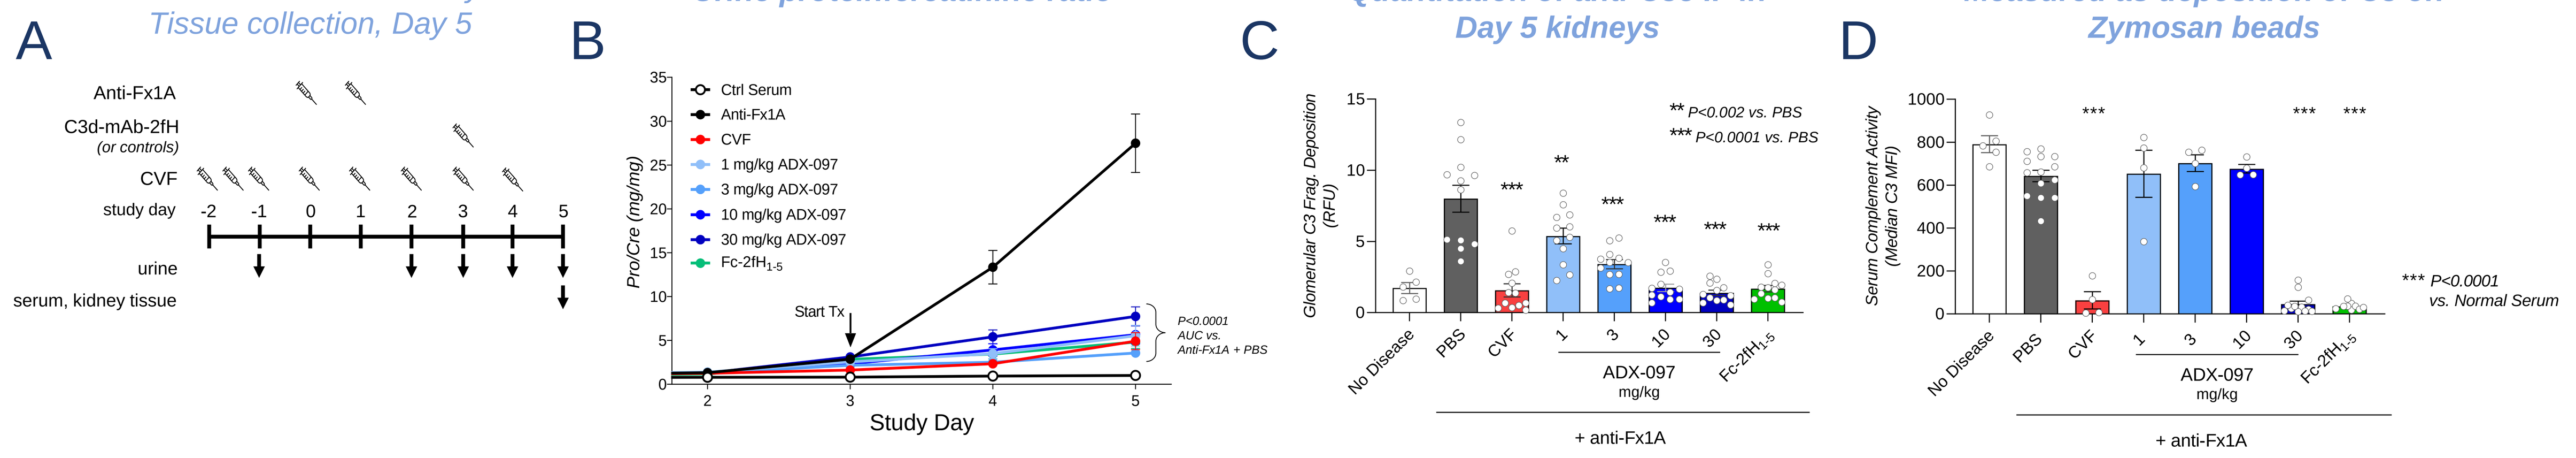
A
Anti-Fx1A
C3d-mAb-2fH
(or controls)
CVF
study day
urine
serum, kidney tissue
B
Ctrl Serum
Anti-Fx1A
CVF
1 mg/kg ADX-097
3 mg/kg ADX-097
10 mg/kg ADX-097
30 mg/kg ADX-097
Fc-2fH1-5
Start Tx
P<0.0001 AUC vs. Anti-Fx1A + PBS
Pro/Cre (mg/mg)
Study Day
C
Glomerular C3 Frag. Deposition (RFU)
** P<0.002 vs. PBS
*** P<0.0001 vs. PBS
No Disease
PBS
CVF
ADX-097 mg/kg
+ anti-Fx1A
D
Serum Complement Activity (Median C3 MFI)
*** P<0.0001 vs. Normal Serum

Figure 2. ADX-097 Ameliorates Proteinuria in Passive Heymann Nephritis. A.) Renal injury is induced by injection of anti-Fx1A serum (sheep serum raised against a rat kidney lysate). Daily treatment with cobra venom factor (CVF) started two days prior to anti-Fx1A treatment. ADX-097 was delivered intravenously (IV) on Day 3, after onset of proteinuria, at the indicated doses. Tissue and Serum was collected on Day 5. **B.)** All tested doses of ADX-097 inhibited progression of proteinuria. The effect was comparable to the CVF-treatment group. **C.)** Kidneys were collected on Day 5 and immunostained with an antibody recognizing active C3 fragments (anti-C3c). C3 fragment deposition was measured by quantifying glomerular immunofluorescence on digital images (≥ 10 glomeruli from each animal on study). ADX-097 treatment dose-dependently reduced glomerular anti-C3c deposition. **D).** Serum was collected on Day 5 from each rat. Complement activity was measured by incubating serum with zymosan particles and measuring deposition of C3 on the particles by flow cytometry. Despite significant inhibition of complement activity in glomerular (panel C), ADX-097 doses ≤ 10 mg/kg showed no inhibition of serum complement.

Study Design:
Disease induction on Day 0
ADX-097 treatment on Day 3
Tissue collection, Day 7

Potent Inhibition of Proteinuria Relies on Tissue Targeting of ADX-097
Non-targeted $Fc\text{-}fH_{1-5}$ fusion is less potent than ADX-097

ADX-097-Mediated Inhibition of Proteinuria Correlates with Preservation of Podocyte Ultrastructure
SEM images of glomeruli collected on Day 7

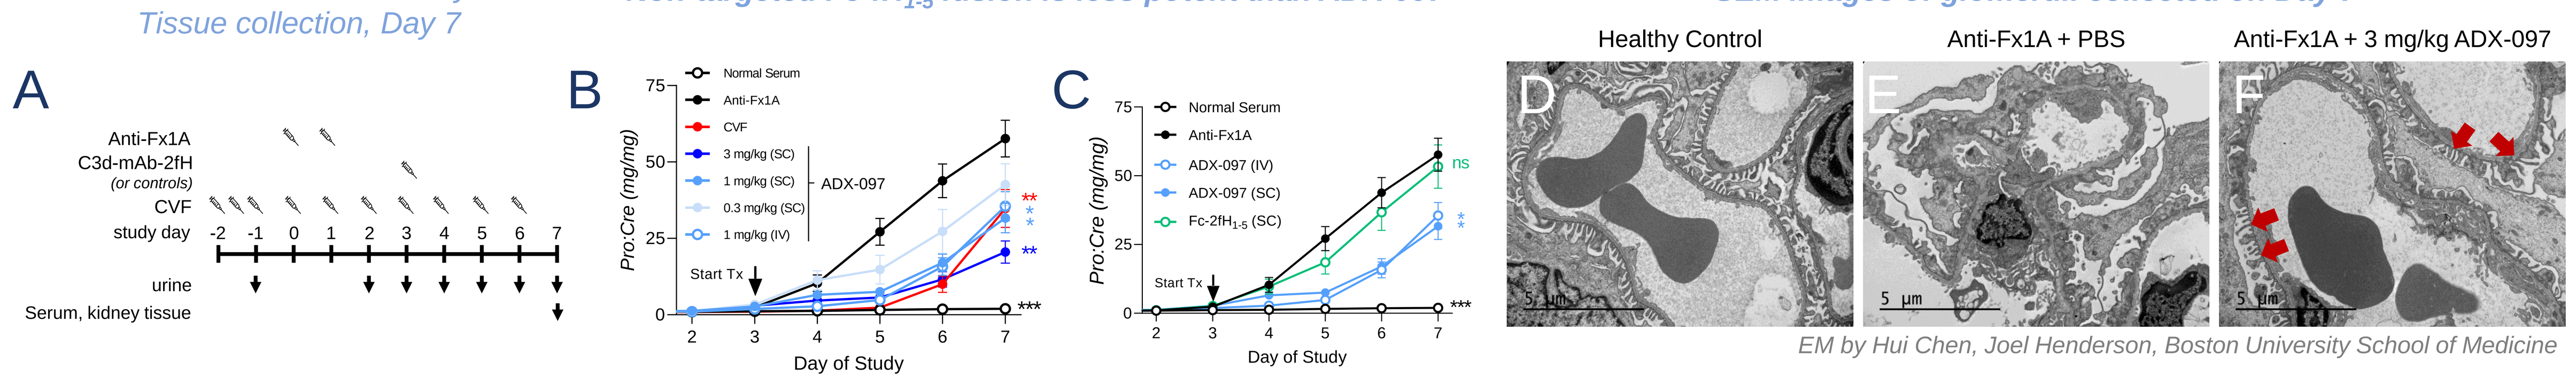
A
Anti-Fx1A
C3d-mAb-2fH
(or controls)
CVF
study day
urine
Serum, kidney tissue
B
Normal Serum
Anti-Fx1A
CVF
3 mg/kg (SC)
1 mg/kg (SC)
0.3 mg/kg (SC)
1 mg/kg (IV)
ADX-097
Start Tx
Pro:Cre (mg/mg)
Day of Study
C
Normal Serum
Anti-Fx1A
ADX-097 (IV)
ADX-097 (SC)
Fc-2fH1-5 (SC)
ns
Healthy Control
Anti-Fx1A + PBS
Anti-Fx1A + 3 mg/kg ADX-097
D
E
F
EM by Hui Chen, Joel Henderson, Boston University School of Medicine

Figure 3. ADX-097 Dose-Dependently Ameliorates Proteinuria in Passive Heymann Nephritis. A.) Lower doses of ADX-097 were evaluated in a Passive Heymann Nephritis study that was performed as above except that ADX-097 was delivered subcutaneously (SC) and tissue and serum was collected on Day 7. **B.)** ADX-097 dose-dependently inhibited progression of proteinuria. **C.)** Comparison of 1 mg/kg IV or SC doses of ADX-097 to an equimolar dose of non-targeted $Fc\text{-}fH_{1-5}$ demonstrates the role of anti-C3d tissue targeted in increasing drug potency. **D).** EM of healthy control rats reveals well-differentiated podocyte foot processes along the urinary side of the glomerular basement membrane (GBM). **E).** EM of Anti-Fx1A + PBS glomeruli shows extensive foot process effacement. **F.)** Treatment of anti-Fx1A-treated rats with ADX-097 shows substantial preservation of podocyte foot processes (red arrows).

Tissue and Circulating PK/PD Characterization of ADX-118, a Mouse Surrogate of ADX-097, in factor H Deficient ($CfH^{-/-}$) Mice

ADX-118 localizes to Glomeruli and Durably Inhibits Complement in $CfH^{-/-}$ Mice
ADX-118 detected by α-fH immunofluorescence (IF)
Complement detected by α-C3 fragment immunofluorescence (IF)

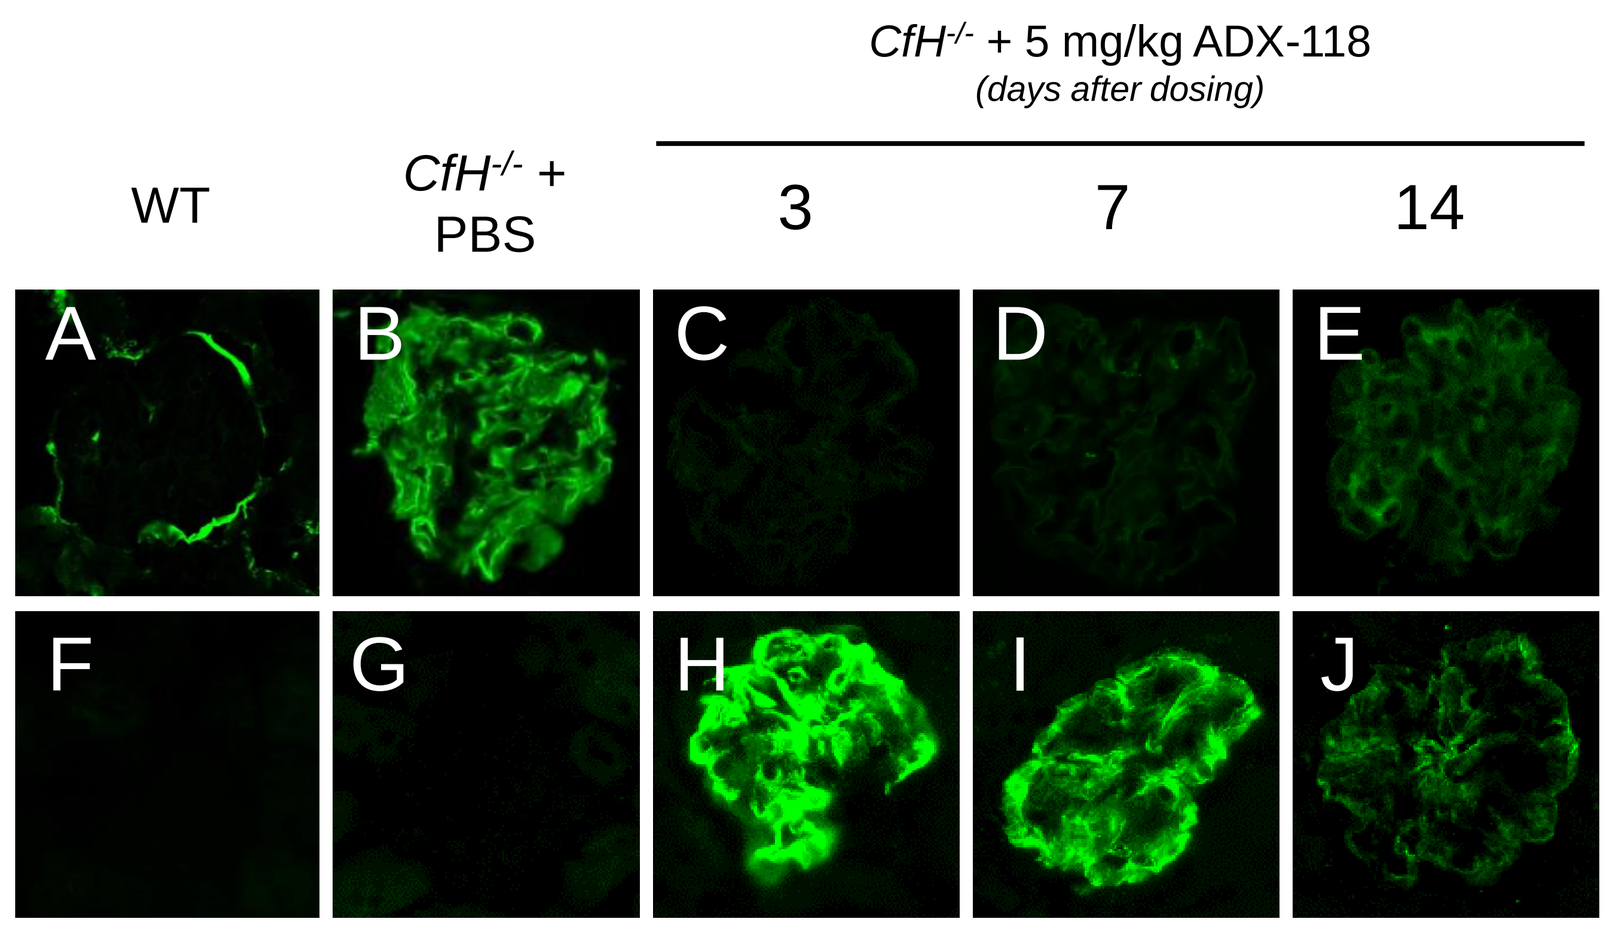
CfH-/- + 5 mg/kg ADX-118
(days after dosing)
WT
CfH-/- + PBS
3
7
14
Complement Activity (anti-C3 frag.)
Drug Exposure (anti-mfH)
A
B
C
D
E
F
G
H
I
J

Figure 4. ADX-118 (mouse surrogate of ADX-097) Localizes to Glomeruli and Potently Inhibits Glomerular Complement in *CfH*$^{-/-}$ mice. A.) Minimal anti-C3 fragment immunofluorescence is detected in WT mice **B.)** anti-C3 fragment immunofluorescence is substantially increased in *CfH*$^{-/-}$ mice (compare with panel A). **C, D, E.)** Delivery of a single dose of ADX-118 inhibits complement activity (measured as anti-C3 fragment immunofluorescence) in *CfH*$^{-/-}$ mice. **F, G.)** No anti-fH immunofluorescence is detected in WT (panel F) or *CfH*$^{-/-}$ mice treated with PBS (panel G). **H, I, J.)** anti-fH immunofluorescence indicates localization of ADX-118 to glomeruli in *CfH*$^{-/-}$ mice. Localization of drug and inhibition of complement are evident for at least two weeks after dosing.

Glomerular and Circulating PK/PD Characterization of ADX-118 in $CfH^{-/-}$ Mice
Kidney: ADX-118 tissue PK detected by α-fH IF • Tissue PD/Complement inhibition detected by α-C3 fragment IF
Circulation (plasma): ADX-118 circulating PK detected by anti-ADX-118 ELISA • Circulating PD/Complement inhibition detected by anti-intact C3 ELISA

Figure 5. Characterization of Tissue and Circulating PK and complement inhibition in *CfH*$^{-/-}$ mice. A single dose of ADX-118 was administered to *CfH*$^{-/-}$ mice at the indicated dose levels. Kidneys and plasma were collected at defined time points (n = 3-4 mice per time point). ADX-118 shows dose-dependent localization in kidney tissue (grey lines, panels **A-E**). ADX-118 doses ≥ 1 mg/kg reach a similar maximum level of complement inhibition (black lines, panels **A-E;** grey bar in each panel indicates average complement inhibition on days 1-7 +/- 1 SD in 25 mg/kg group). Consistent with data presented in **Figure 4**, tissue complement inhibition quite durable, with complete inhibition lasting at least a week, and partial inhibition observed 12-14 days after ADX-118 dosing. Because intact circulating C3 protein is ~20x lower in *CfH*$^{-/-}$ mice than in WT mice, inhibition of circulating complement activity can be measured by assaying intact C3 protein levels in plasma. As illustrated in panels **F** and **J**, sufficiently high circulating concentrations of ADX-118 lead to a transient increase in plasma intact C3 that reflects inhibition of systemic complement, and this effect disappears as ADX-118 clears from circulation. However, subcutaneous delivery of ≤ 5 mg/kg ADX-118 yields no measurable increase in plasma intact C3 (panels **G, H, I**). Taken together, these data indicate that 1-5 mg/kg doses of ADX-118 potently and durably inhibit tissue complement without affecting systemic complement

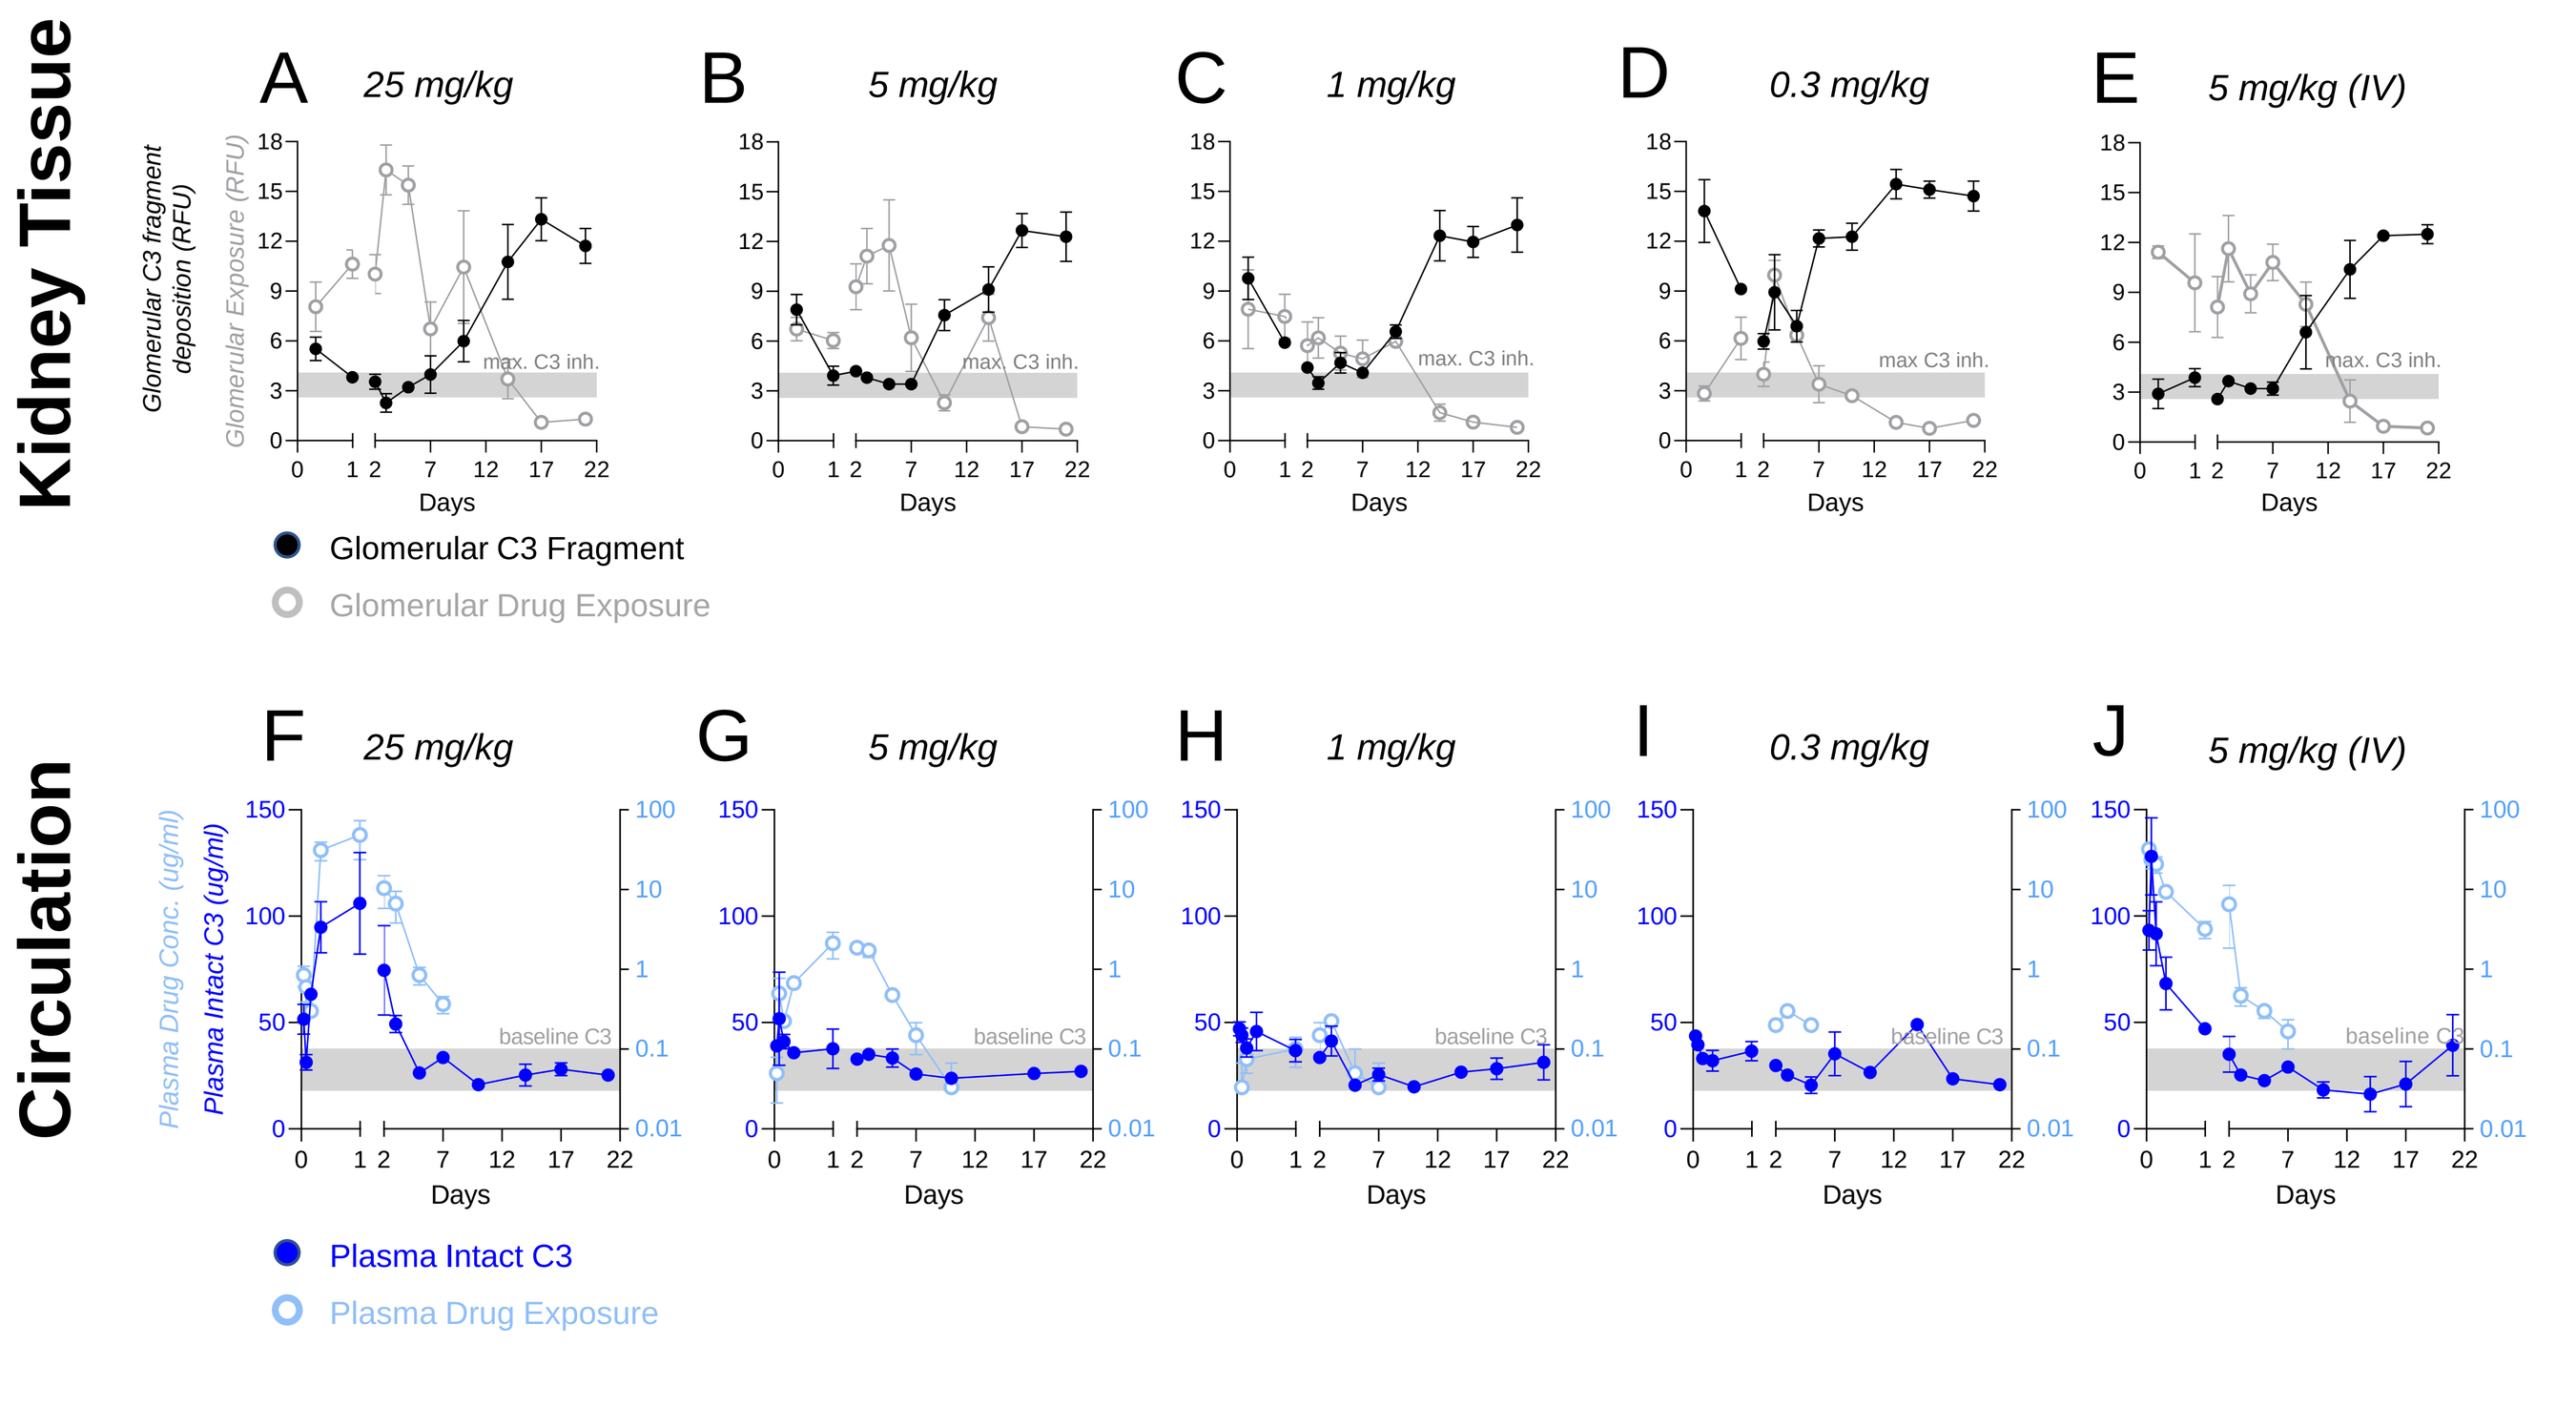
Kidney Tissue
A 25 mg/kg
B 5 mg/kg
C 1 mg/kg
D 0.3 mg/kg
E 5 mg/kg (IV)
Glomerular C3 fragment deposition (RFU)
Glomerular Exposure (RFU)
max. C3 inh.
Days
Glomerular C3 Fragment
Glomerular Drug Exposure
Circulation
F 25 mg/kg
G 5 mg/kg
H 1 mg/kg
I 0.3 mg/kg
J 5 mg/kg (IV)
Plasma Drug Conc. (ug/ml)
Plasma Intact C3 (ug/ml)
baseline C3
Plasma Intact C3
Plasma Drug Exposure

ADX-097 Delivers Local Complement Inhibition to Injured Primate Skin

Study Design:
Skin UVB exposure on Day -1
ADX-097 treatment on Day 0
Daily biopsy collection, Days 1-6

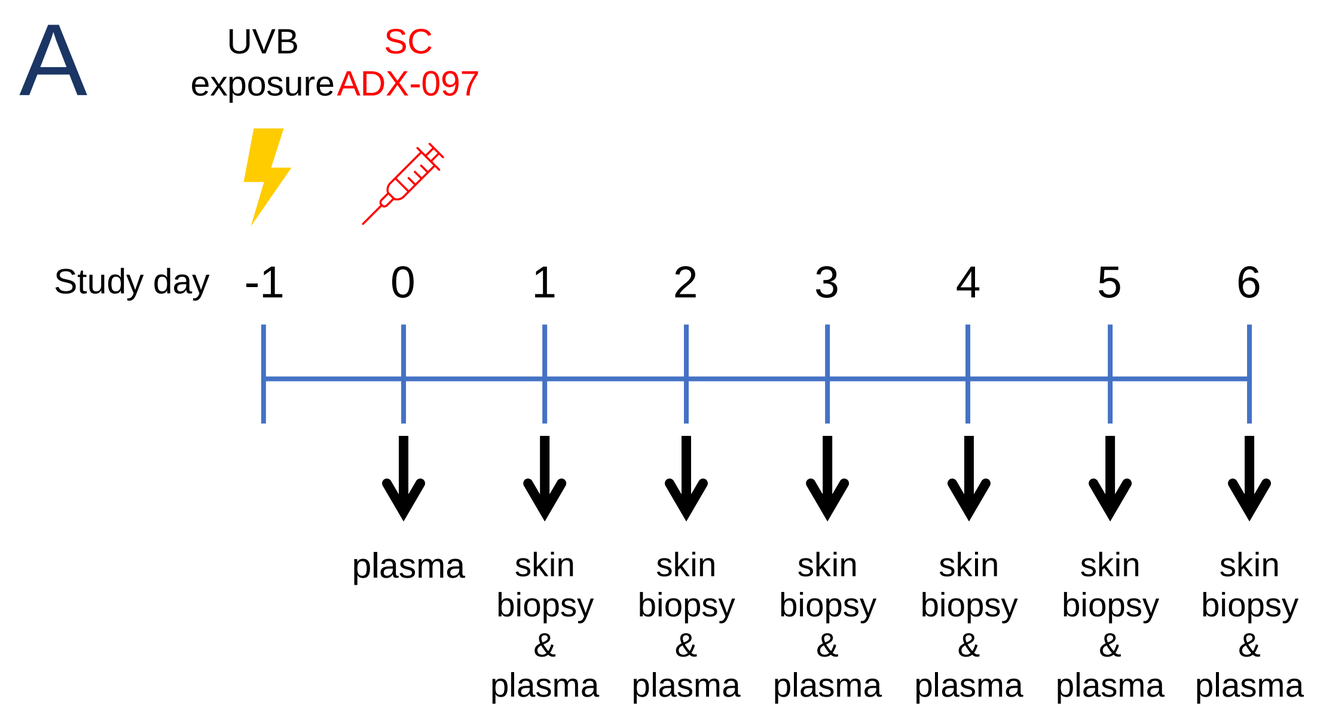
A
UVB exposure
SC ADX-097
Study day
-1
0
1
2
3
4
5
6
plasma
skin biopsy & plasma

ADX-097 Locally Inhibits Complement in Injured Primate Skin
***ADX-097** - anti-fH IF*
Skin Complement – anti-C3 fragments (C3c)

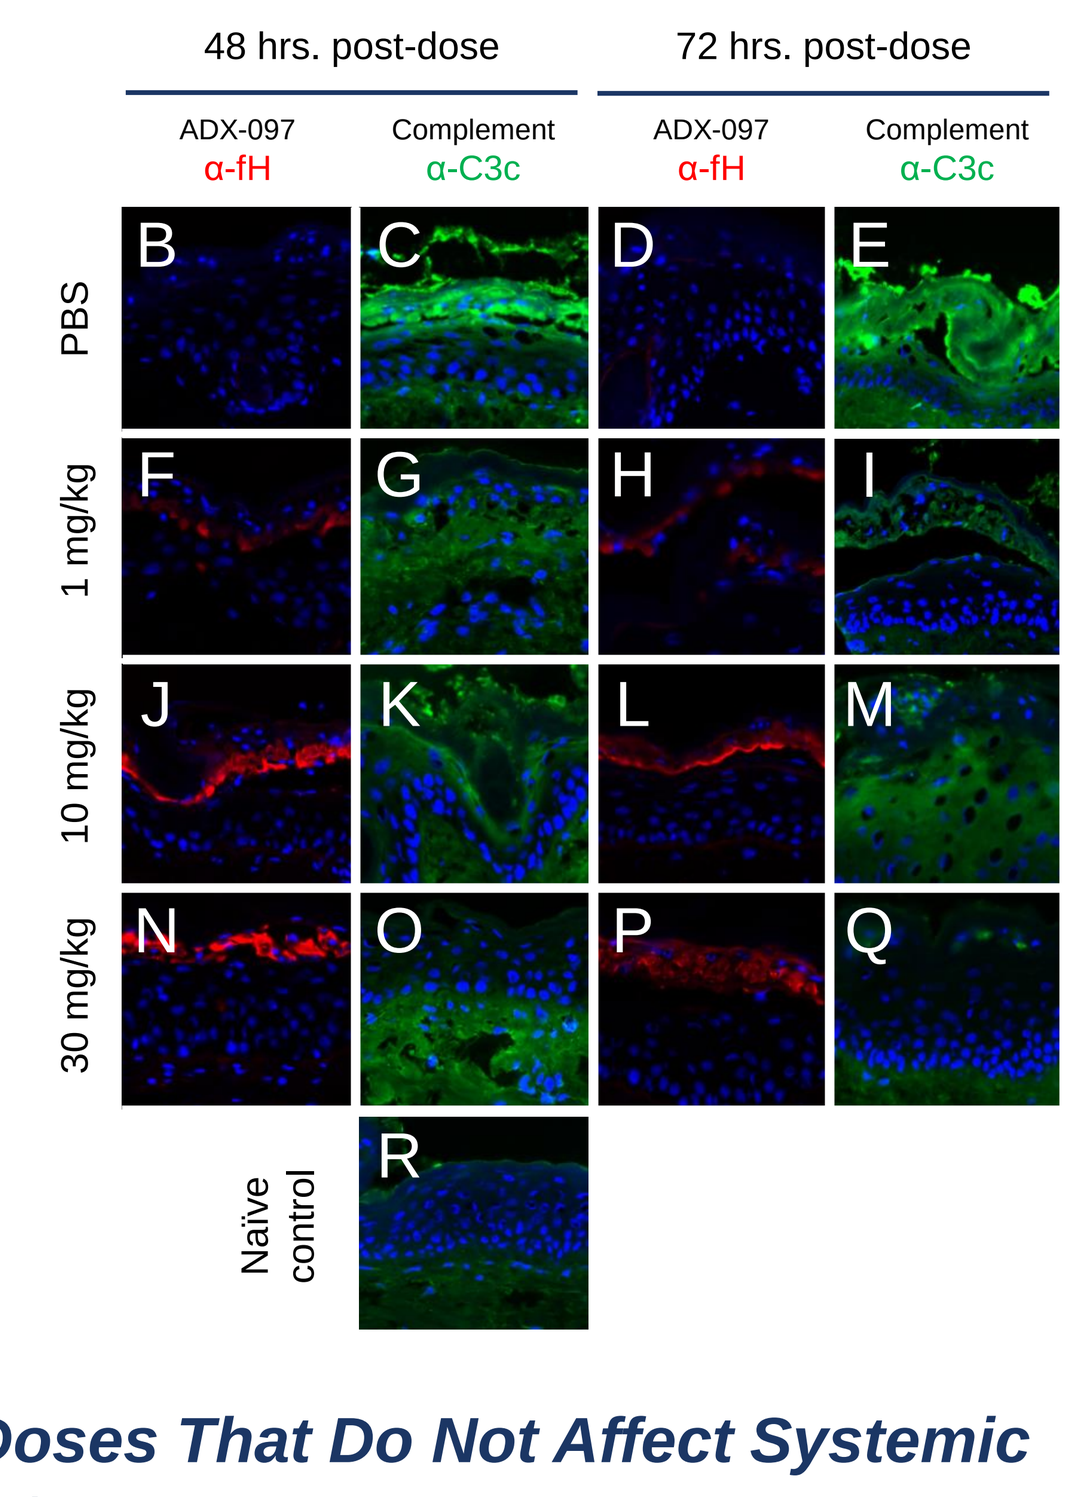
48 hrs. post-dose
72 hrs. post-dose
ADX-097 α-fH
Complement α-C3c
PBS
1 mg/kg
10 mg/kg
30 mg/kg
Naive control

Figure 6. ADX-097 Locally Inhibits Complement in Injured Skin. A.) Complement was induced in Cynomolgus monkey skin by exposing shaved skin to 3 MED UVB light. 24 hours later, ADX-097 was delivered SC (at a site distant from UVB exposure). Skin biopsies were collected every 24 hours for 6 days after ADX-097 dosing. ADX-097 can be detected in injured skin (compare **B** to **F, J,** and **N** and **D** to **H, L,** and **P).** Complement is induced after UVB exposure (compare **C** and **E** to **R**). Presence of ADX-097 in skin correlates with reduced skin complement (compare **C** to **G, K, O** and **E** to **I, M, Q**).

ADX-097 Inhibits NHP Skin Complement at Doses That Do Not Affect Systemic Complement
Skin: ADX-097 PK (α-fH) • Tissue complement (α-C3 fragment)
Circulation (plasma): ADX-097 PK (ELISA) • Systemic complement (Wieslab assay)

Figure 7. ADX-097 Tissue and Circulating PK and Complement Inhibition. A.) 10 and 30 mg/kg doses of ADX-097 completely inhibit systemic complement (measured in plasma by Wieslab assay). 1 mg/kg ADX-097 minimally inhibits systemic complement. **B.)** All tested doses of ADX-097 equivalently inhibit skin complement after UVB exposure. Skin complement measured by image quantitation of anti-C3 fragment immunofluorescence. **C.)** ADX-097 shows dose-dependent plasma exposure (measured by ADX-097-specific ELISA). Loss of systemic complement inhibition correlates with ADX-097 circulating exposures below ~75 μg/ml. Maximum circulating exposure in the ADX-097 1 mg/kg group is < 10 μg/ml. **D.)** Drug localization to UVB-exposed skin was measured by image quantitation of anti-fH immunofluorescence. ADX-097 localizes to skin in a dose-dependent manner. Note that this is in contrast to inhibition of skin complement (panel **B**), suggesting that full saturation of local C3d is not required for maximum ADX-097 activity.

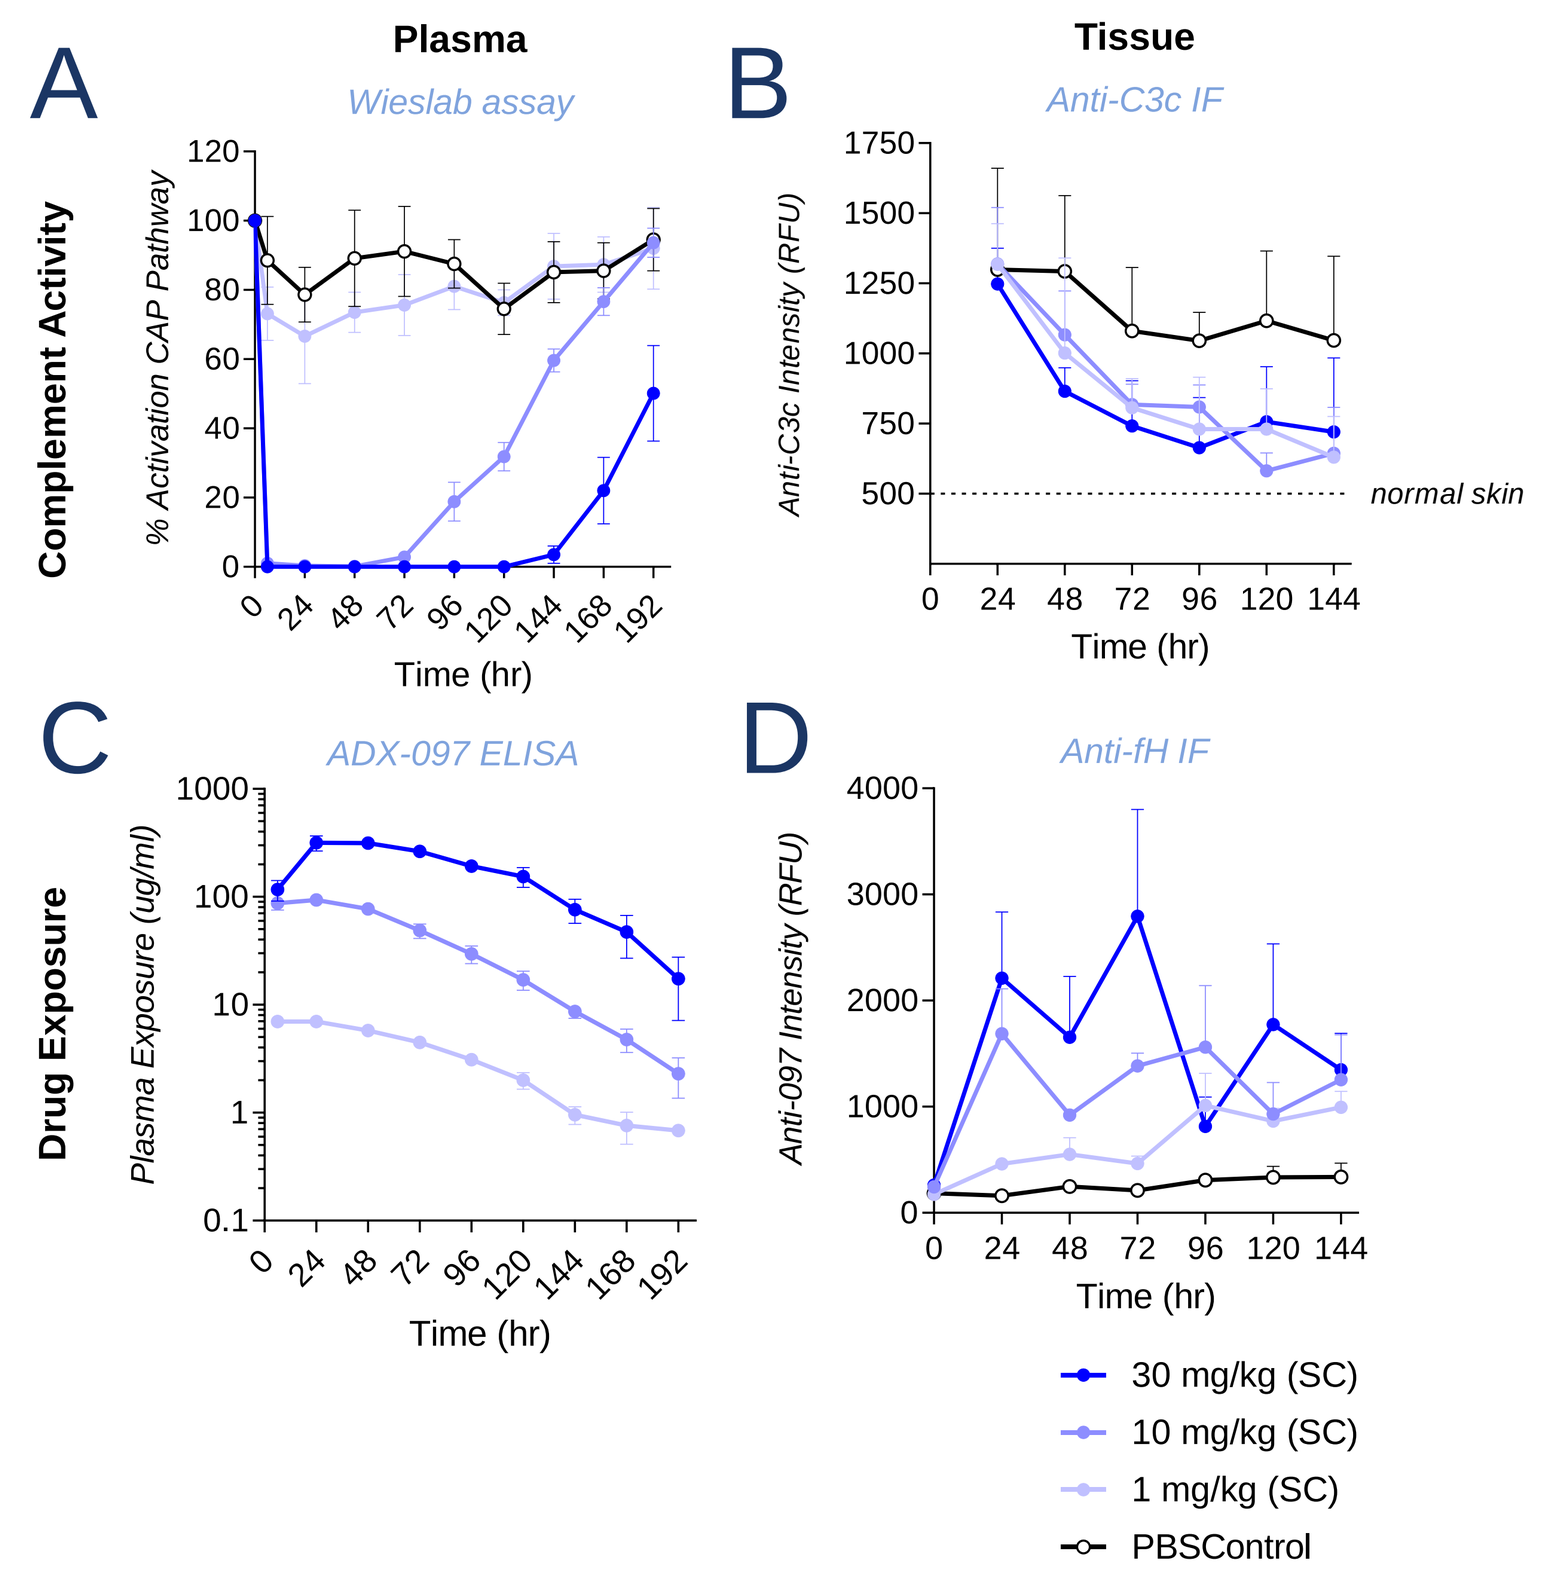
Plasma
Tissue
A
Wieslab assay
Complement Activity
% Activation CAP Pathway
Time (hr)
B
Anti-C3c IF
Anti-C3c Intensity (RFU)
normal skin
C
ADX-097 ELISA
Drug Exposure
Plasma Exposure (ug/ml)
D
Anti-fH IF
Anti-097 Intensity (RFU)
30 mg/kg (SC)
10 mg/kg (SC)
1 mg/kg (SC)
PBSControl

C3d Deposition is a Common Feature of Glomerular Disease, Suggesting Broad Clinical Applicability for C3d-Targeted Complement Inhibition

C3d Deposition in Human Glomerular Disease
anti-C3d immunofluorescence

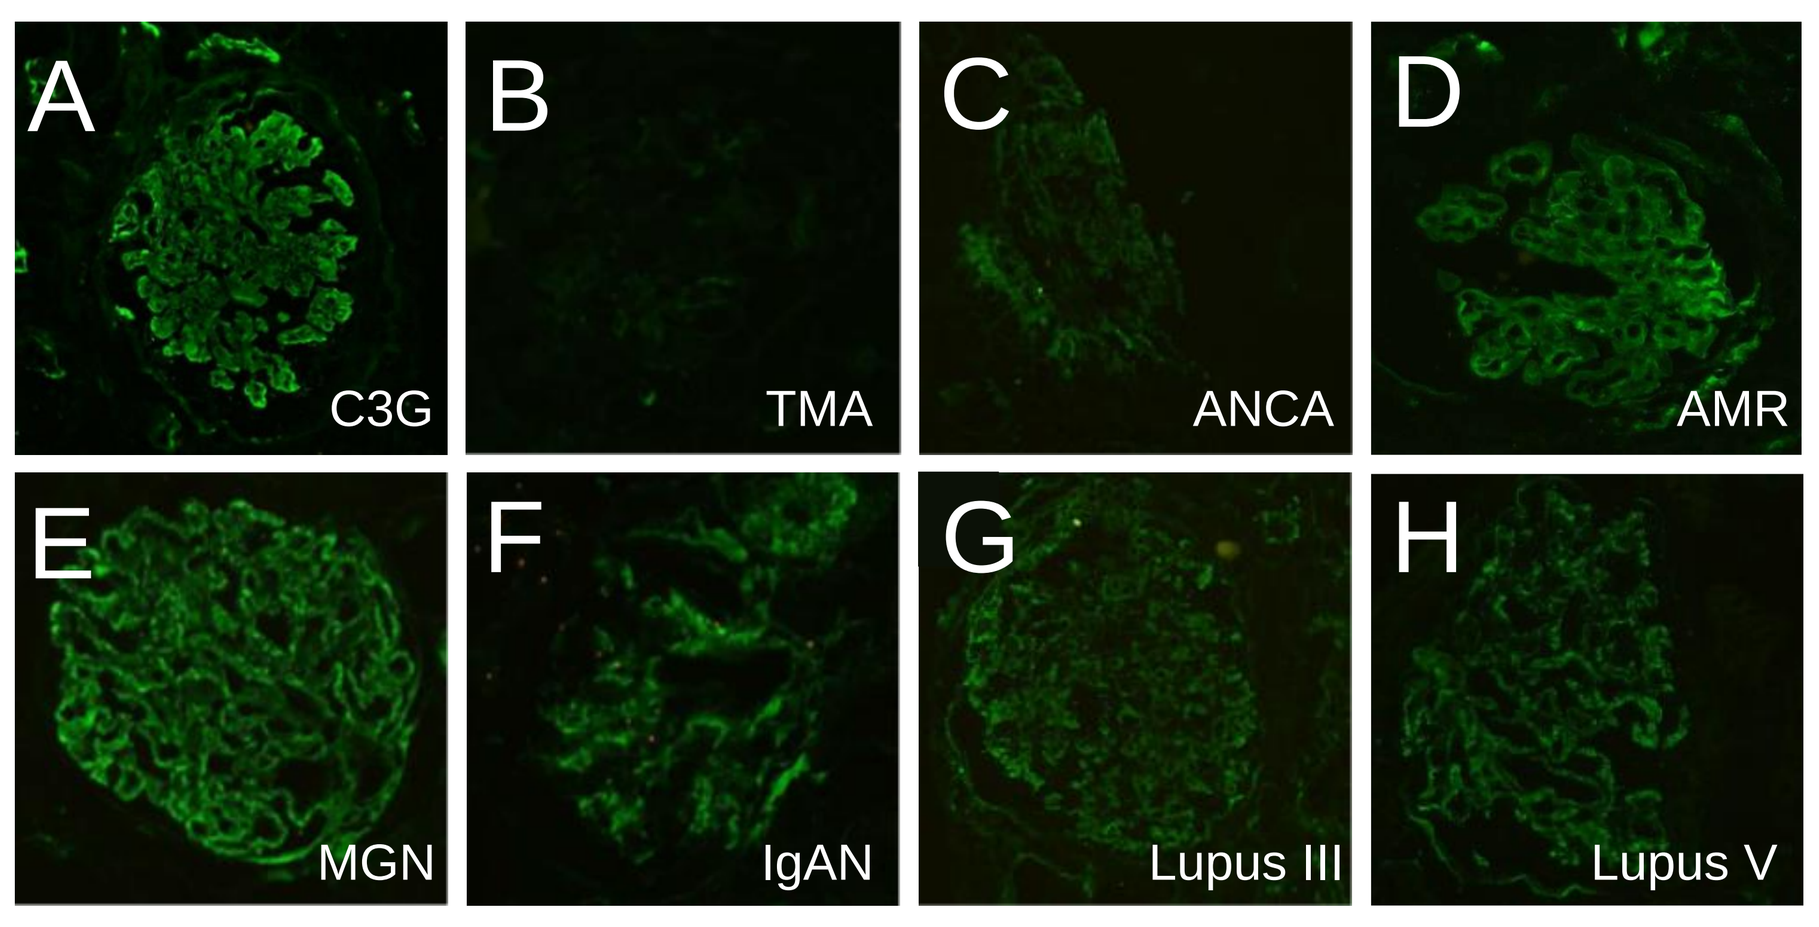
A C3G
B TMA
C ANCA
D AMR
E MGN
F IgAN
G Lupus III
H Lupus V

Semi-Quantitative Scoring of anti-C3d and anti-C3 Fragment Immunofluorescence in Human Glomerular Disease

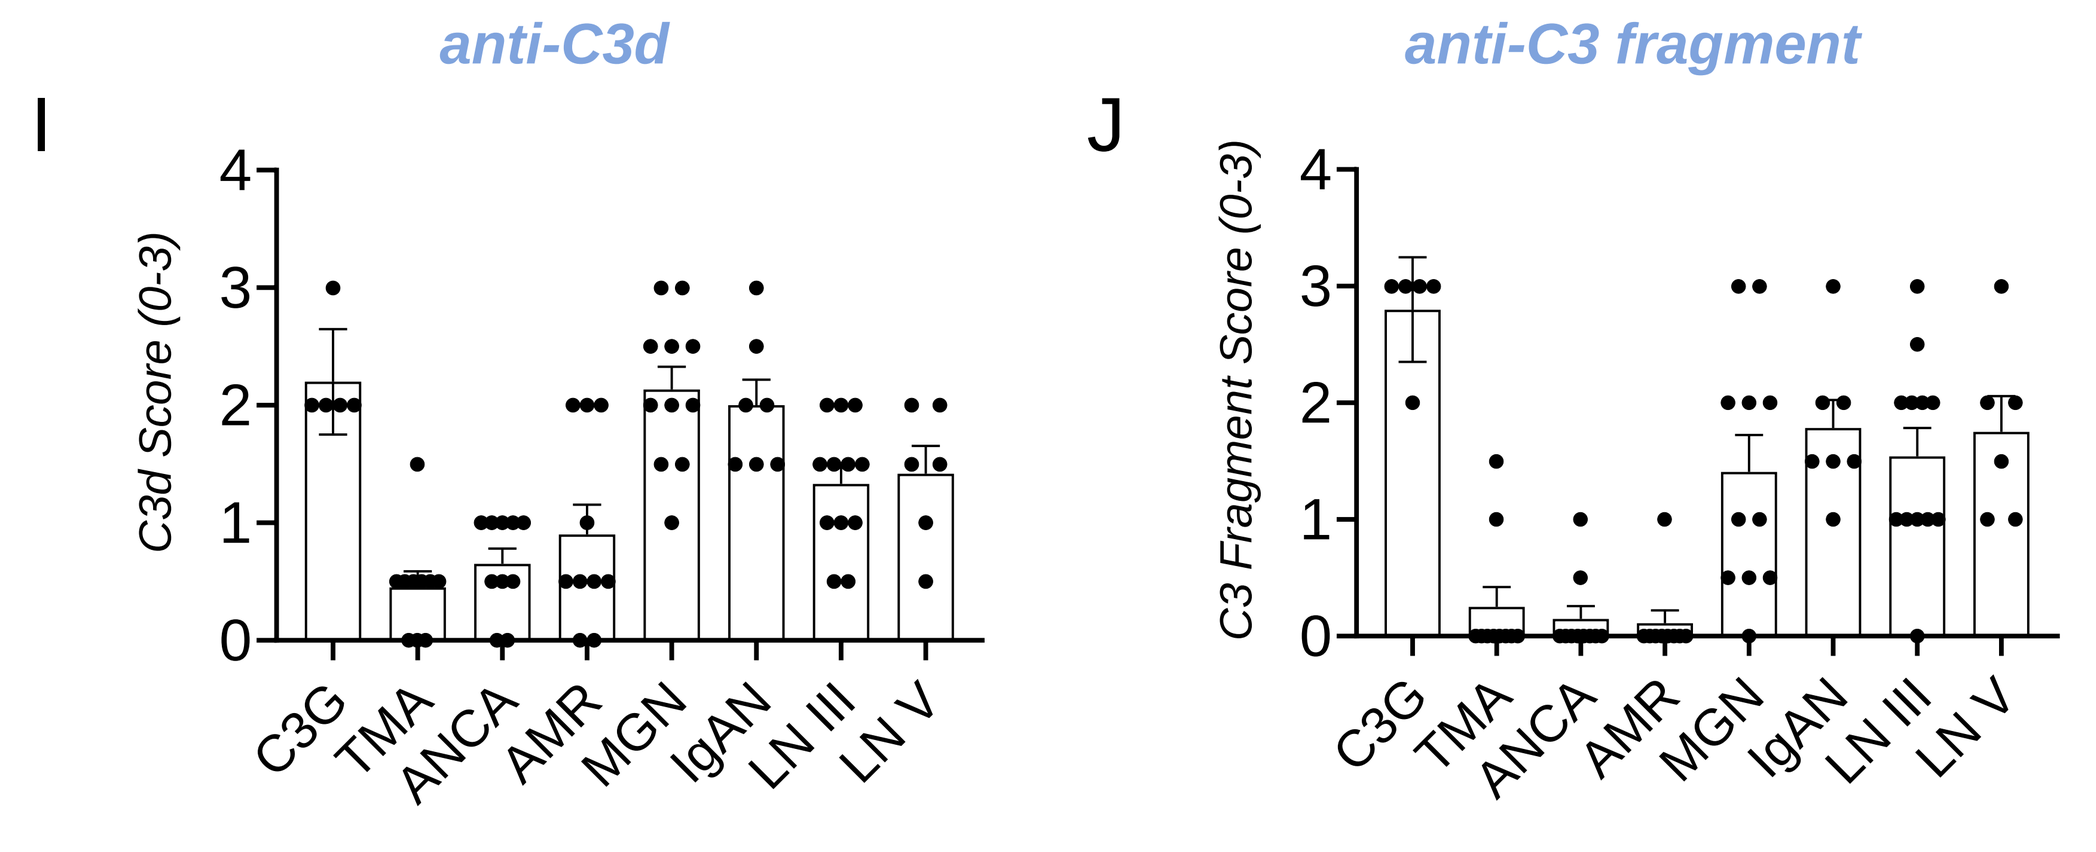
anti-C3d
anti-C3 fragment
I
J
C3d Score (0-3)
C3 Fragment Score (0-3)
C3G
TMA
ANCA
AMR
MGN
IgAN
LN III
LN V

Figure 8. C3d and C3 Fragment Deposition in Human Glomerular Disease Biopsies. Panels **A-H.)** Anti-C3d immunofluorescence in glomeruli from **A.)** C3 Glomerulopathy (C3G) **B.)** Thrombotic microangiopathy (TMA) **C.)** ANCA vasculitis **D.)** Antibody-mediated rejection of transplant (AMR) **E).** Membranous glomerulopathy (MGN) **F.)** IgA Nephropathy (IgAN), and Class III **(G)** and Class IV **(H)** Lupus. **I.)** anti-C3d immunostaining intensity in all samples was scored by a pathologist, confirming generally higher C3d deposition in MGN, IgAN, and both classes of lupus nephritis. **J.)** These findings are also broadly consistent with similar quantitation of anti-C3 fragment staining from the same human biopsies

Summary & Conclusions

- ADX-097 and its mouse surrogate, ADX-118, are antibody fusion proteins that target complement inhibition to sites of tissue complement activity
- ADX-097 reduces proteinuria and restores podocyte foot process architecture in the Passive Heyman Nephritis (PHN) model of kidney injury
 - Anti-C3d targeting improves the potency of ADX-097
 - ADX-097 is efficacious at doses that do not inhibit systemic complement
- PK/PD characterization of ADX-118 in $CfH^{-/-}$ mice demonstrates potent, durable glomerular complement inhibition at doses that do not affect systemic complement
- Characterization of ADX-097 in a cynomolgus monkey model of skin complement confirms mouse PK/PD, demonstrating potent, durable complement inhibition without affecting systemic complement in primates
- C3d deposition is a common feature of complement-mediated glomerular injury, indicating the potential of broad therapeutic applicability of a C3d-targeted complement inhibitor